Antisense oligonucleotide therapeutic candidates targeting regulatory RNAs to treat urea cycle disorders

Bryan Matthews, Yun Joon Jung, Gabby Bedard, Eric Coughlin, Brynn Akerberg, Rachana Kelkar, Preeti Sathe, Henry Moukheiber, Shraddha Kamath, Amy Jiang, Brian Schwartz, Jiaqi Huang, Abeer Almutairy, Yuting Liu, Jesse Lugus, Shubh Roy, Yuchun Guo, John Miller, Alla Sigova, Alfica Sehgal, and David Bumcrot

CAMP4 Therapeutics Corp., Cambridge, MA, USA

Abstract

Diseases caused by haploinsufficiencies or hypomorphic mutations could be potentially addressed by elevating expression of the healthy alleles of the affected disease-associated genes. Even relatively modest increases (less than 2-fold) can be highly impactful on disease phenotype. Regulatory RNAs (regRNAs), including enhancer RNAs and promoter-associated RNAs, are key regulators of gene transcription. Our RAP Platform enables both the identification of regRNAs associated with a target gene and upregulation of the target gene's expression using antisense oligonucleotides (ASOs). Here, we describe how we are applying this approach to develop and characterize therapeutic product candidates targeting regRNAs associated with the human ornithine transcabamylase gene (OTC) for the potential treatment of Urea Cycle Disorders (UCDs) such as OTC deficiency (OTCD).

Using our RAP Platform, we identified key cis-elements regulating the transcription of OTC gene in human and mouse hepatocytes and candidate regRNAs transcribed from these cis-elements. We then designed and screened ASOs targeting the OTC regRNAs and discovered several ASOs that specifically upregulated OTC mRNA in human hepatocytes in vitro in a dose-dependent manner. We further demonstrated the efficacy of the selected ASOs in a humanized mouse model, in which ASO treatment resulted in OTC upregulation, significant reduction in plasma ammonia levels as well as comcommitant increase in urea levels. To investigate the mechanism by which the selected ASOs induce OTC gene expression upregulation, we evaluated ASO target engagement activities and their ability to modify the secondary structure of regRNAs. We show that selected ASOs modulated the binding of negative transcriptional regulators resulting in increased OTC transcription.

Our results support the therapeutic potential of ASOs targeting OTC regRNAs for the treatment of UCDs.

Background

Ornithine Transcarbamylase Deficiency (OTCD) is an X-linked urea cycle disorder resulting in an inability to adequately process ammonia to urea

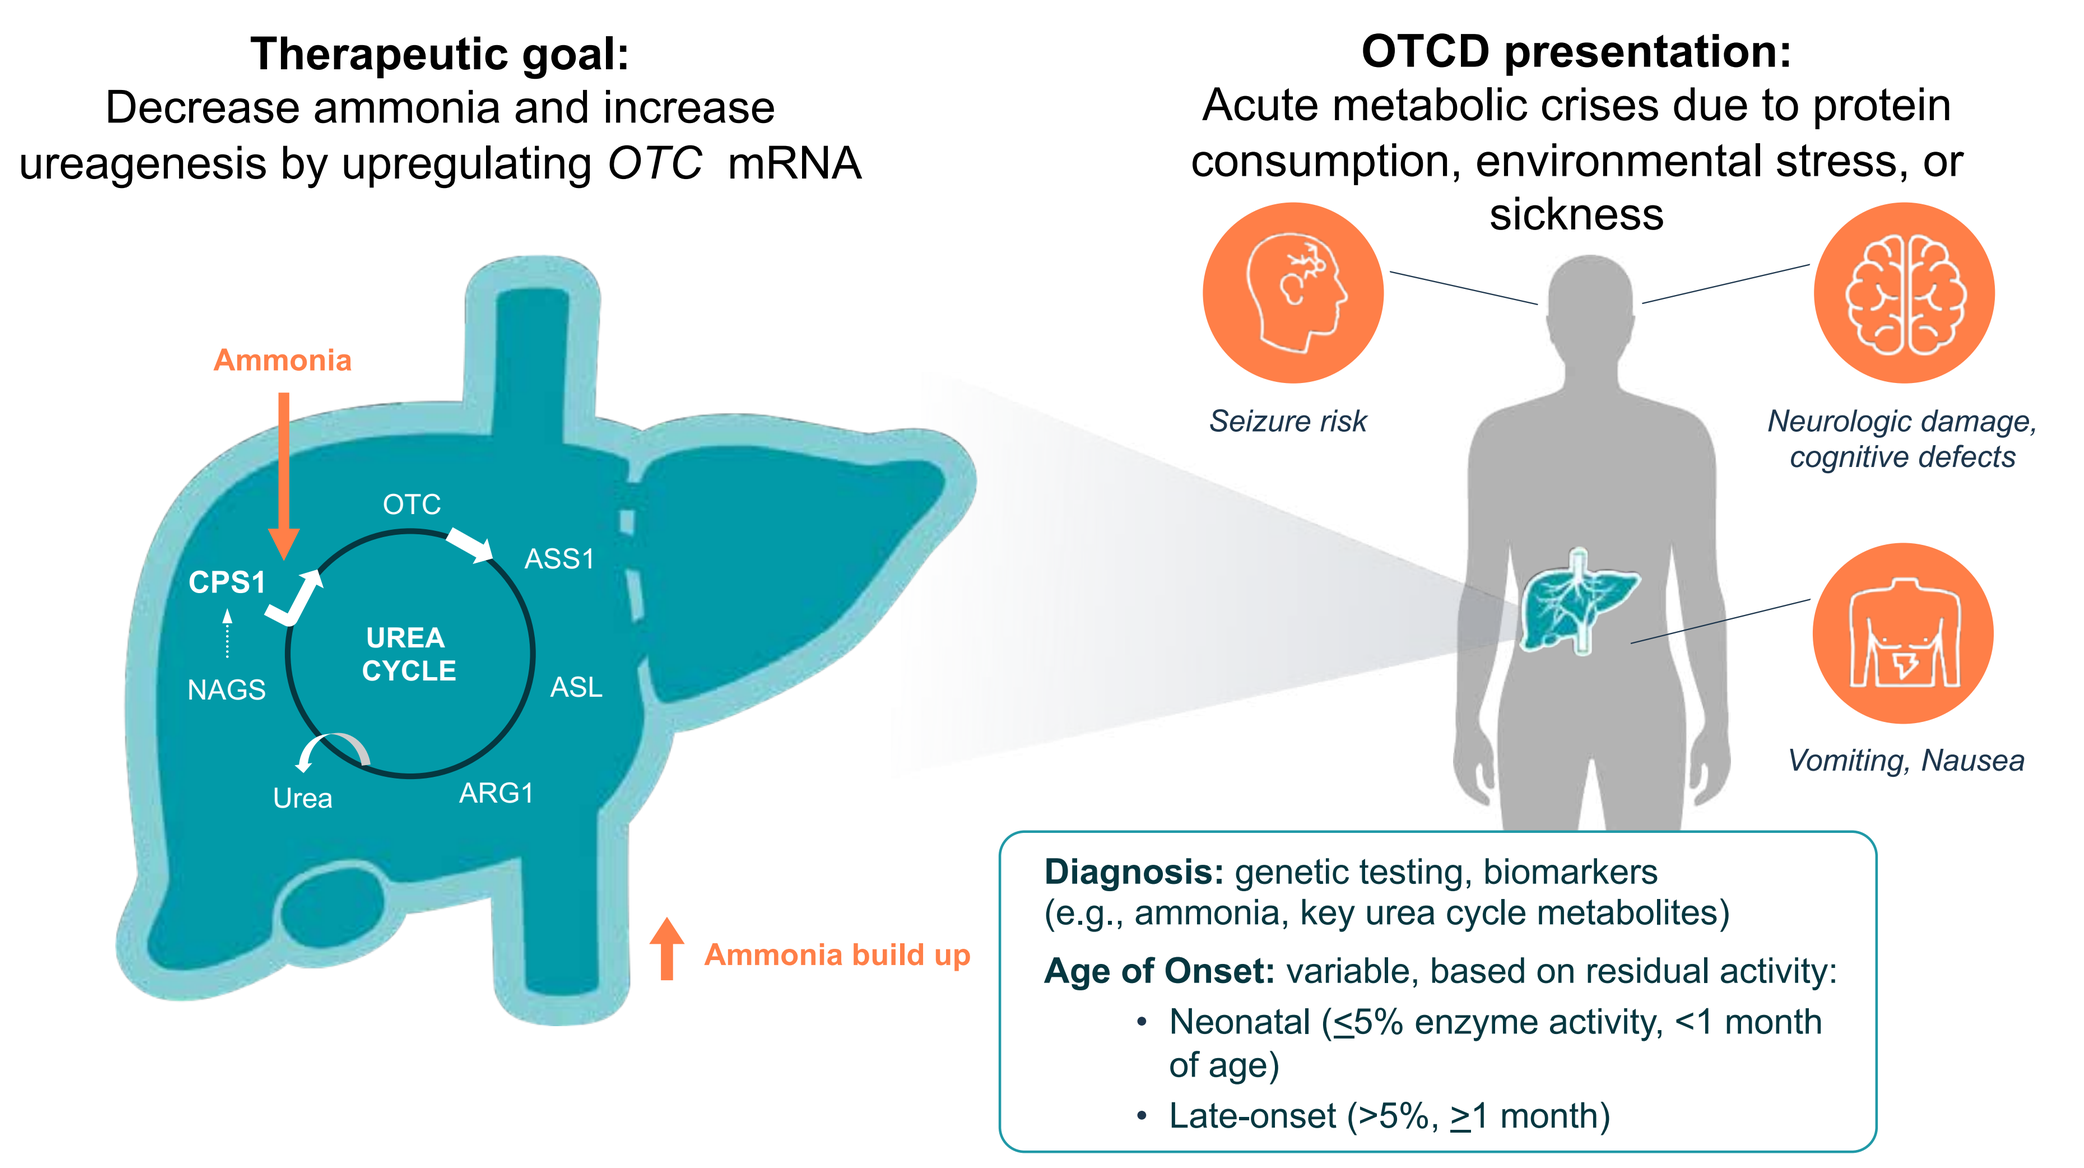
Therapeutic goal:
Decrease ammonia and increase ureagenesis by upregulating OTC mRNA
OTCD presentation:
Acute metabolic crises due to protein consumption, environmental stress, or sickness
Ammonia
OTC
CPS1
ASS1
UREA CYCLE
NAGS
ASL
Urea
ARG1
Ammonia build up
Seizure risk
Neurologic damage, cognitive defects
Vomiting, Nausea
Diagnosis: genetic testing, biomarkers (e.g., ammonia, key urea cycle metabolites)
Age of Onset: variable, based on residual activity:
• Neonatal (≤5% enzyme activity, <1 month of age)
• Late-onset (>5%, ≥1 month)

Regulatory RNAs (regRNAs) are a broad class of cis-acting noncoding RNAs

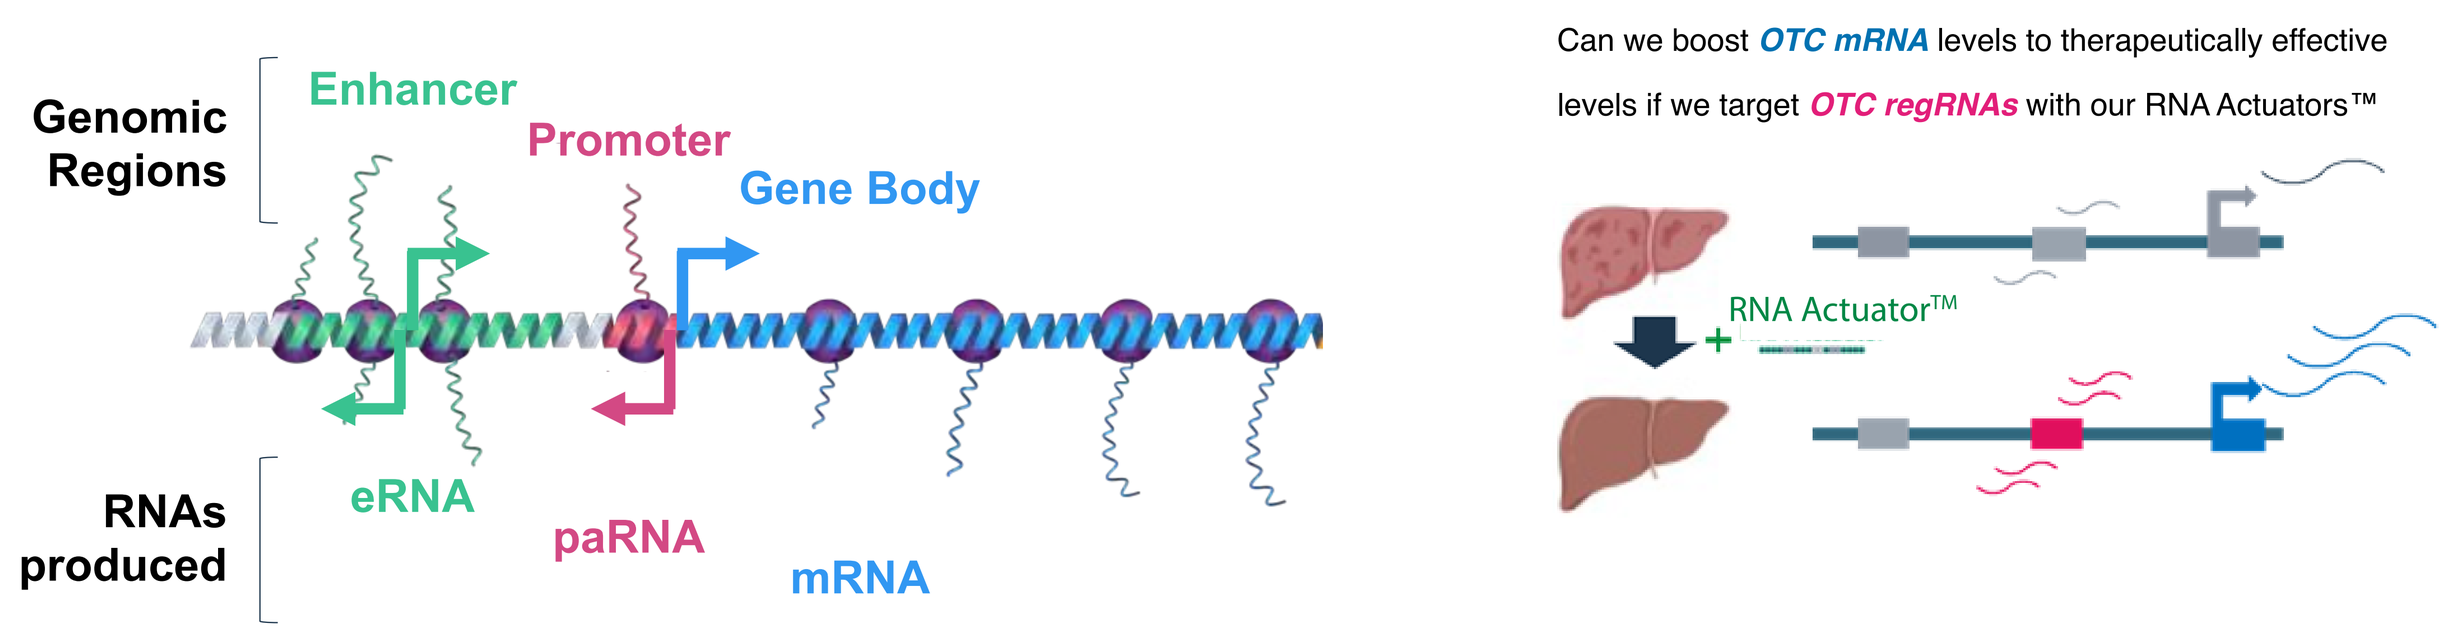
Genomic Regions
Enhancer
Promoter
Gene Body
RNAs produced
eRNA
paRNA
mRNA
Can we boost OTC mRNA levels to therapeutically effective levels if we target OTC regRNAs with our RNA Actuators™
RNA Actuator™

RAP Platform™: Programming medicines targeting regRNAs

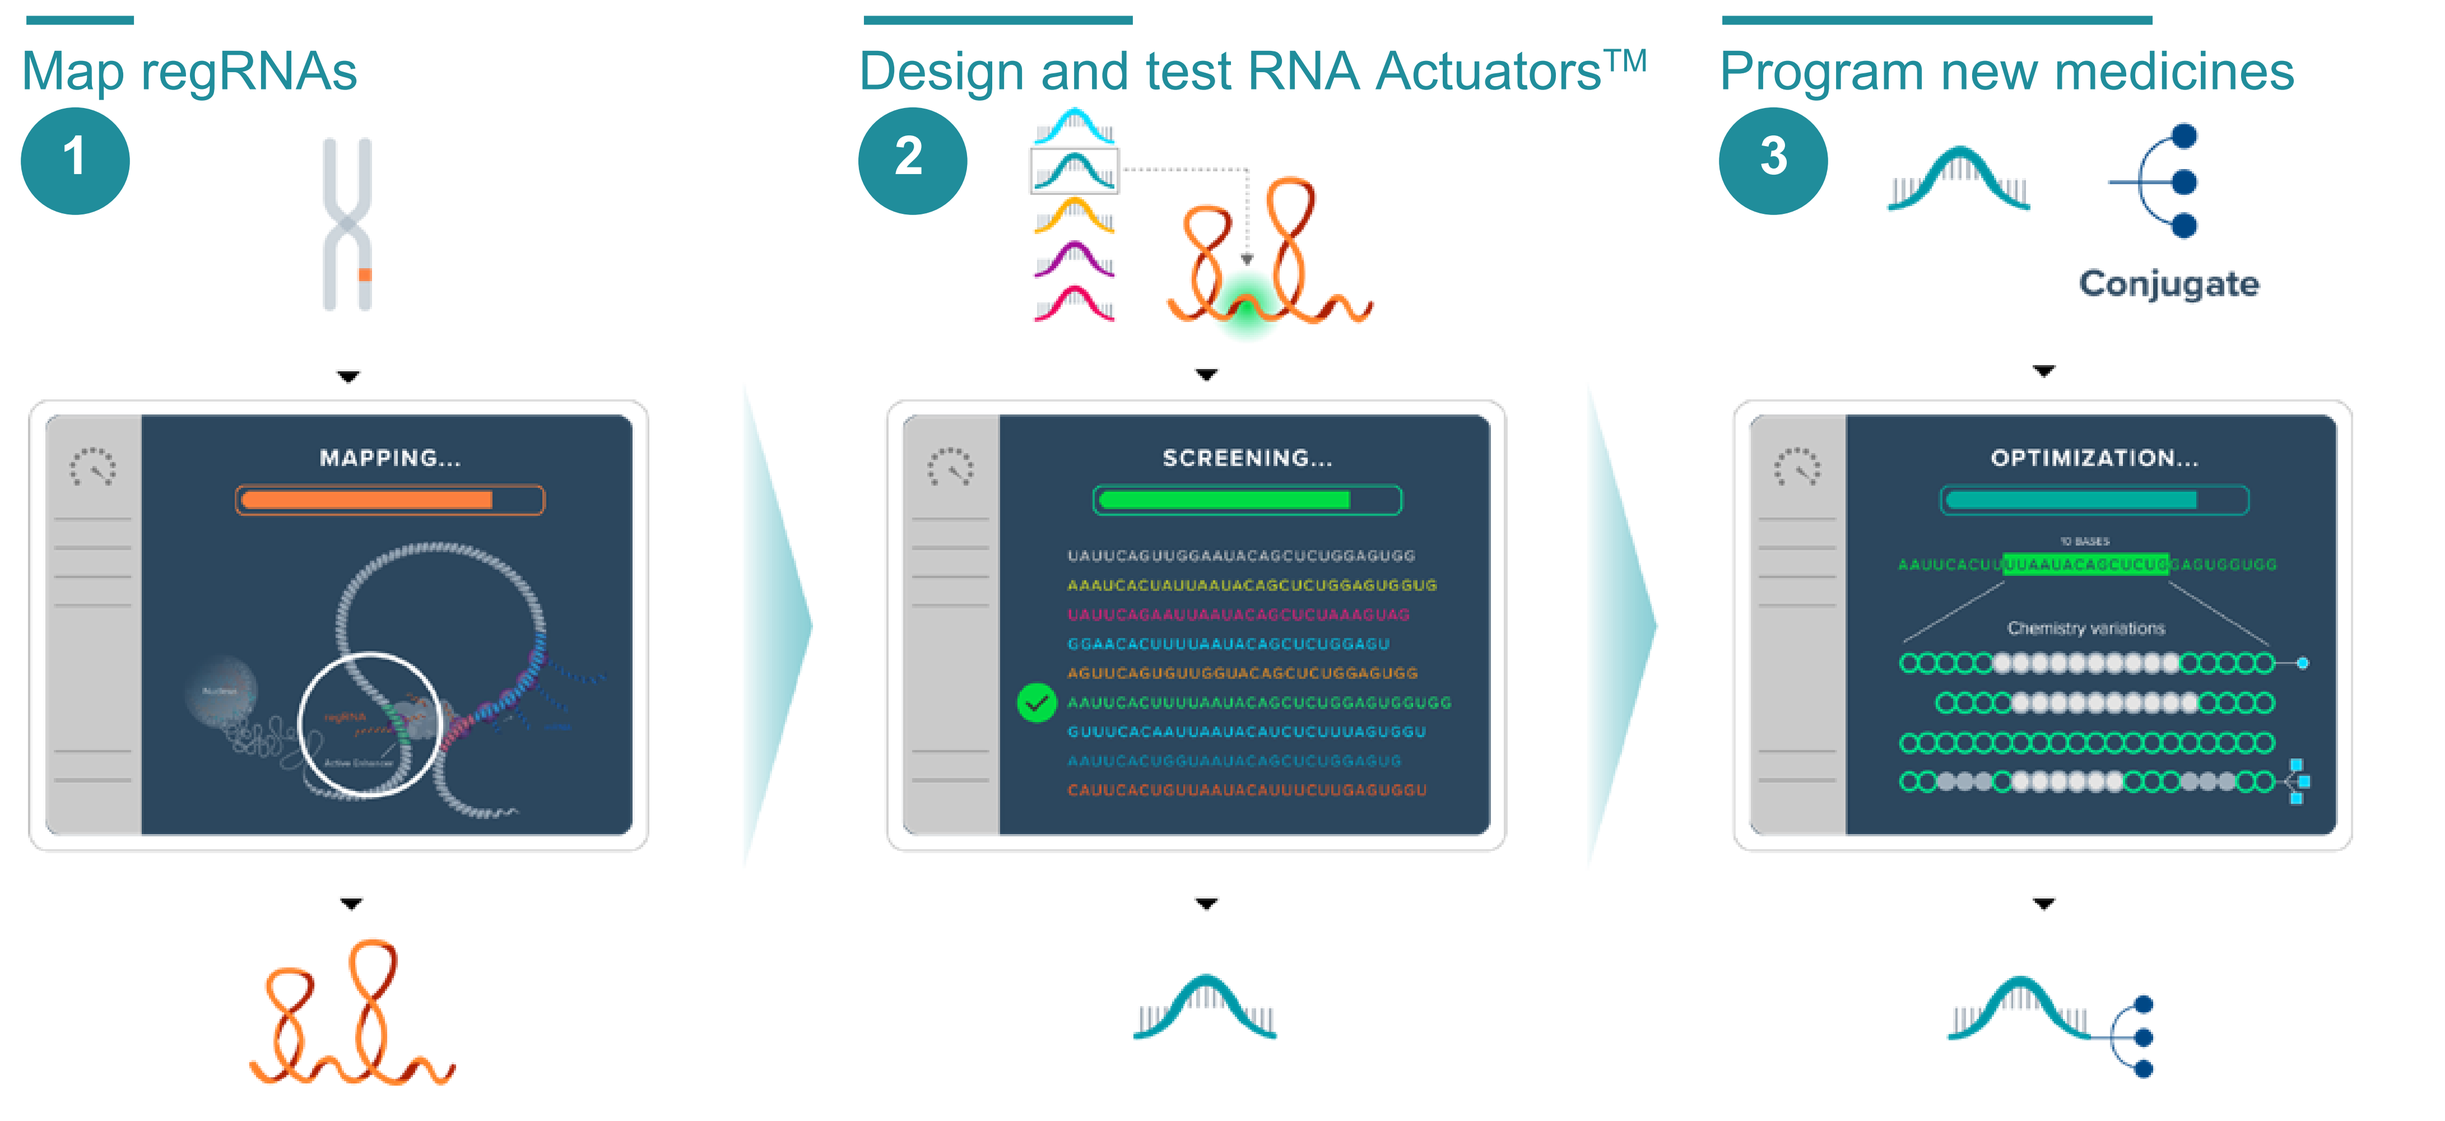
Map regRNAs
1
Design and test RNA Actuators™
2
Program new medicines
3
Conjugate
MAPPING...
SCREENING...
OPTIMIZATION...

Mapping *OTC* locus in primary human hepatocytes

Upstream OTC enhancer is a functionally-validated regulatory region
- OTC enhancer exhibits hallmark open chromatin and histone modification
- regRNA is produced bidirectionally from enhancer
- CRISPRa/i targeting the functional enhancer modulates *OTC* expression

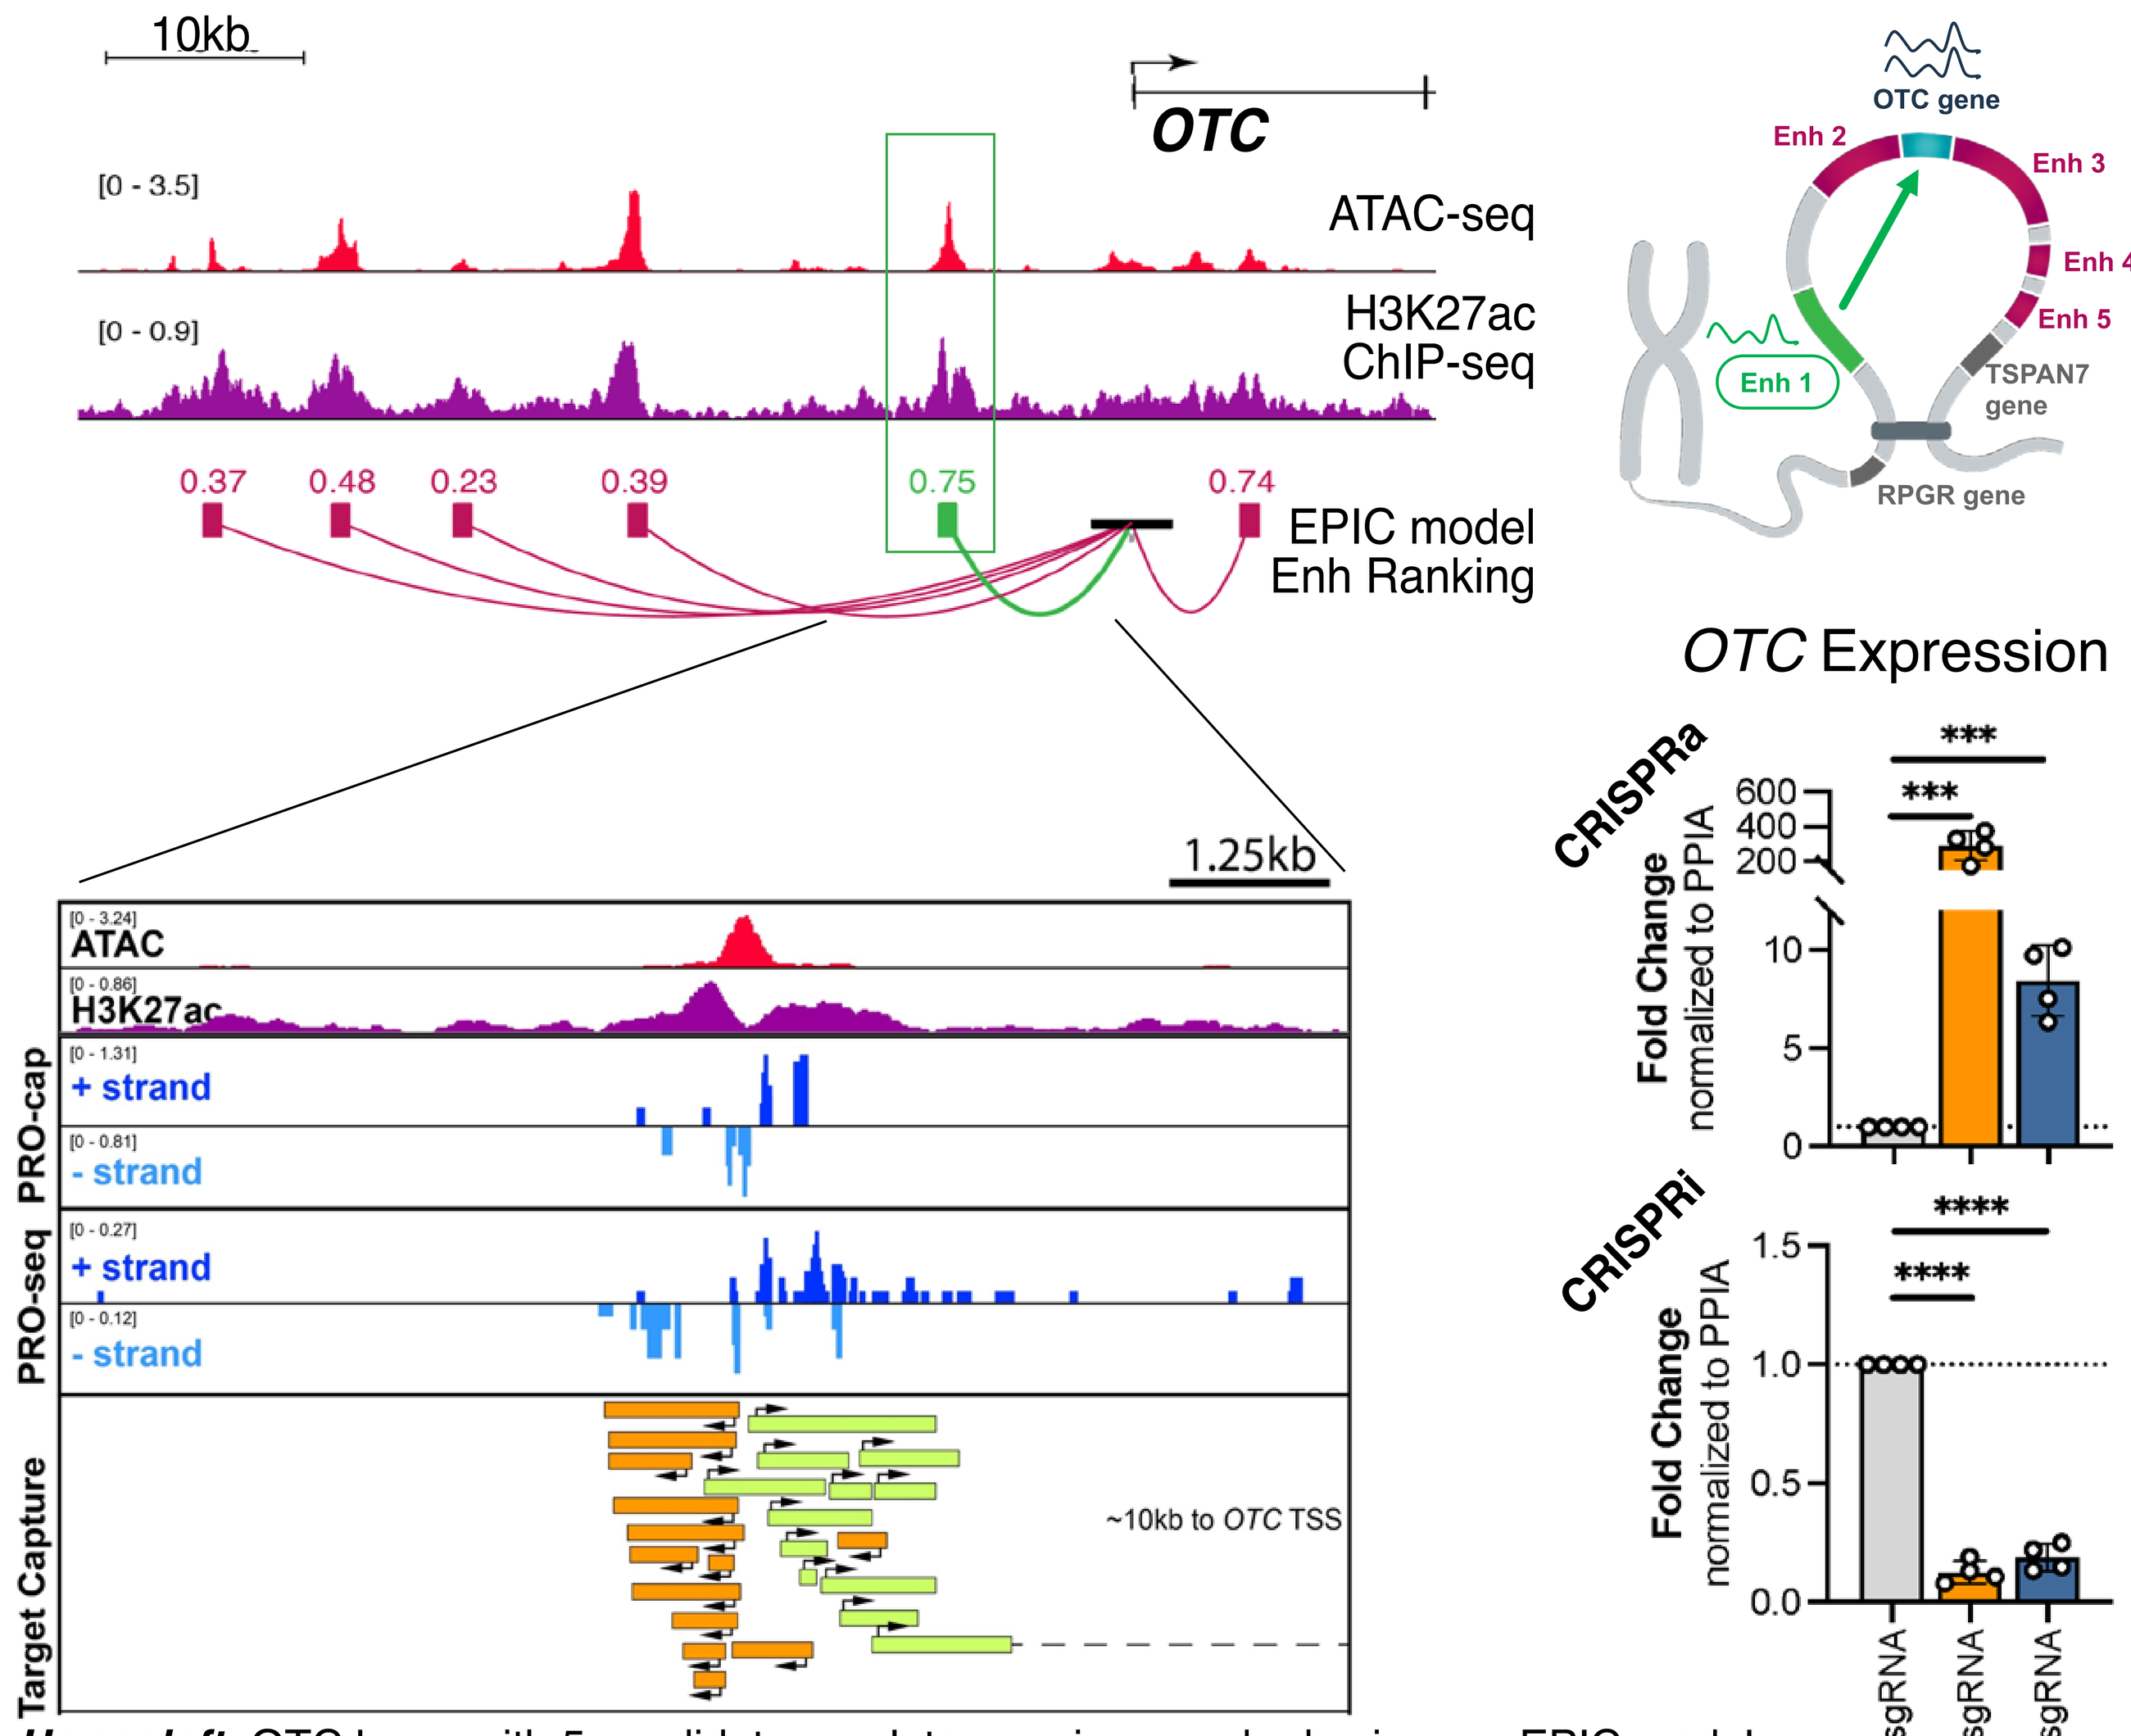
10kb
OTC
[0 - 3.5]
ATAC-seq
[0 - 0.9]
H3K27ac ChIP-seq
0.37
0.48
0.23
0.39
0.75
0.74
EPIC model Enh Ranking
OTC gene
Enh 1
Enh 2
Enh 3
Enh 4
Enh 5
TSPAN7 gene
RPGR gene
1.25kb
ATAC
H3K27ac
PRO-cap
+ strand
- strand
PRO-seq
+ strand
- strand
Target Capture
~10kb to OTC TSS
OTC Expression
CRISPRa
CRISPRi
Fold Change normalized to PPIA

NTC sgRNA
TSS sgRNA
enh sgRNA

Upper left: OTC locus with 5 candidate regulatory regions, ranked using our EPIC model. For more information regarding our EPIC modeling see our other poster (Guo et al.)
Lower left: as +10kb OTC enhancer is transcribed bidirectionally as measured by PRO-seq, PRO-cap, and a target capture methodology to enrich for regRNAs of interest.
Right: Guide RNAs targeting the OTC enhancer in HepG2 cells expressing CRISPRa (VPH) or CRISPRi (KRAB) effectors validate that the enhancer is functional. This effect is specific to OTC (data not shown).

ID and optimization of *OTC* regRNA-targeting ASOs

ASO screening identifies lead dose-responsive ASOs

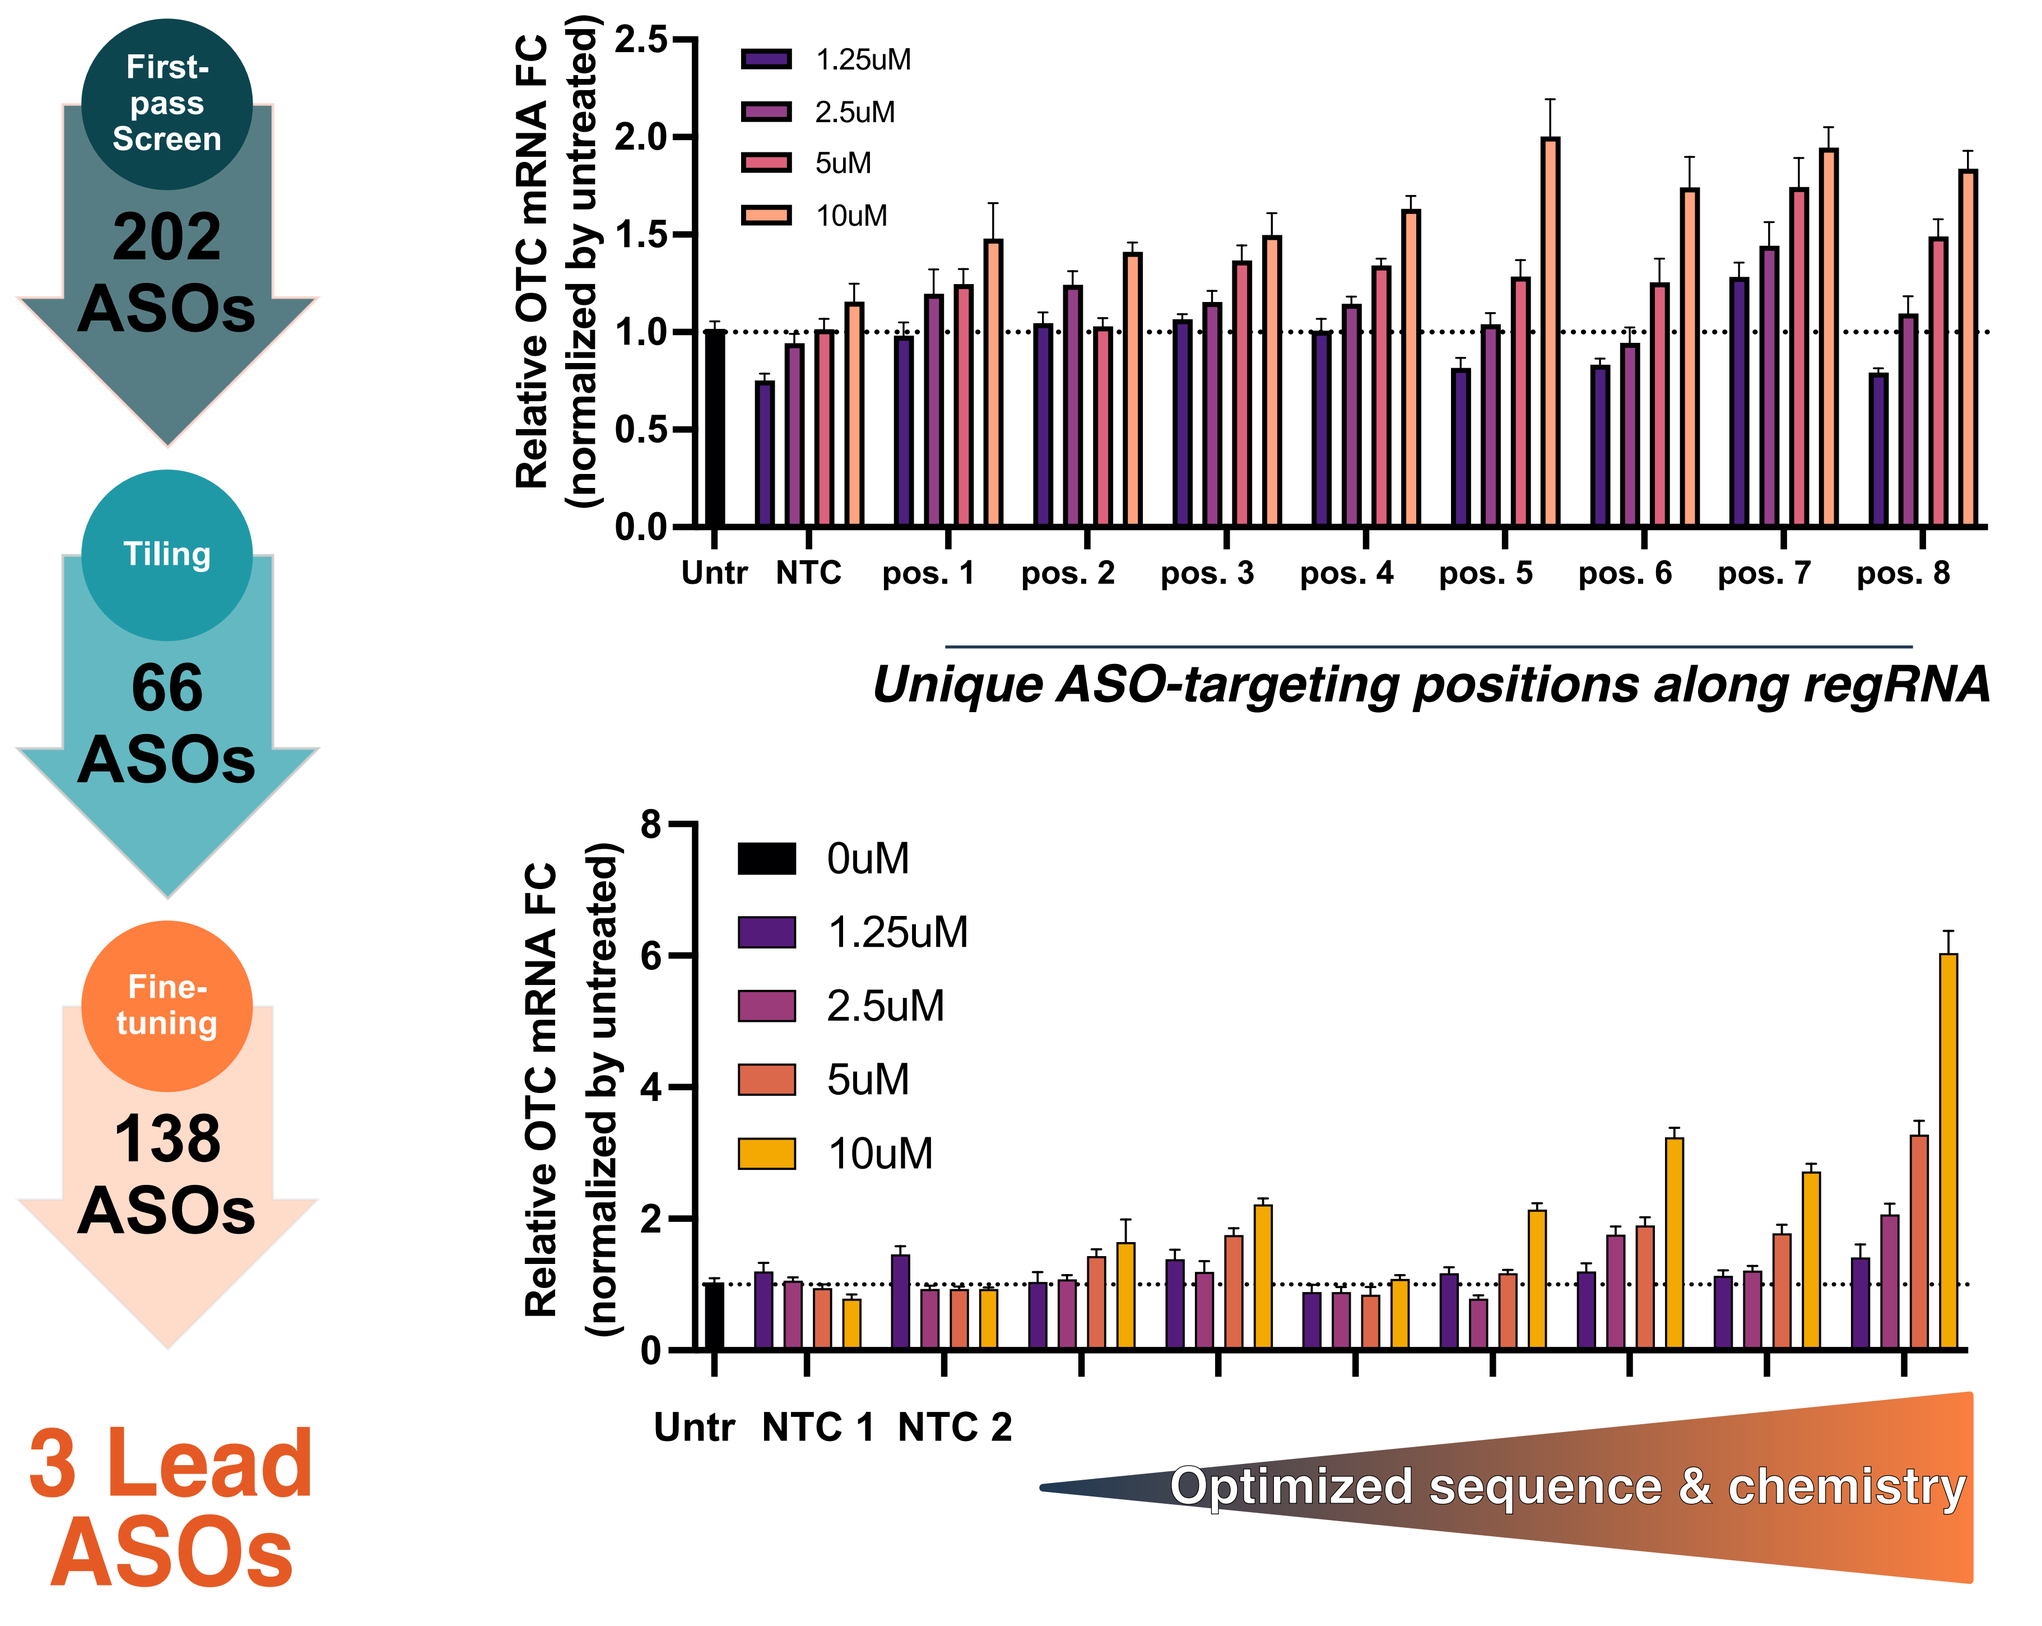
First-pass Screen
202 ASOs
Tiling
66 ASOs
Fine-tuning
138 ASOs
3 Lead ASOs
Relative OTC mRNA FC (normalized by untreated)
1.25uM
2.5uM
5uM
10uM
Untr
NTC
pos. 1
pos. 2
pos. 3
pos. 4
pos. 5
pos. 6
pos. 7
pos. 8
Unique ASO-targeting positions along regRNA
0uM
Untr
NTC 1
NTC 2
Optimized sequence & chemistry

Above: Top Graph- ASO screening of ASOs targeting different positions along the regRNA for their ability to upregulate *OTC* mRNA, resulting in multiple hits with >1.5 Fold-change (FC). ***Bottom graph-*** Optimization of lead ASO chemistry and sequence improves its ability to upregulate OTC mRNA in vitro.
Below: Treatment with an ASO targeting the minus strand regRNA results in upregulation of the target *OTC* regRNA within 2h followed by a later upregulation of *OTC* mRNA at 12 and 24h.
All: FC by quantitative or digital PCR relative to a housekeeper gene (HPRT or PPIA, as indicated) and sample indicated on the y-axis

Upregulation of targeted regRNA precedes *OTC* mRNA changes

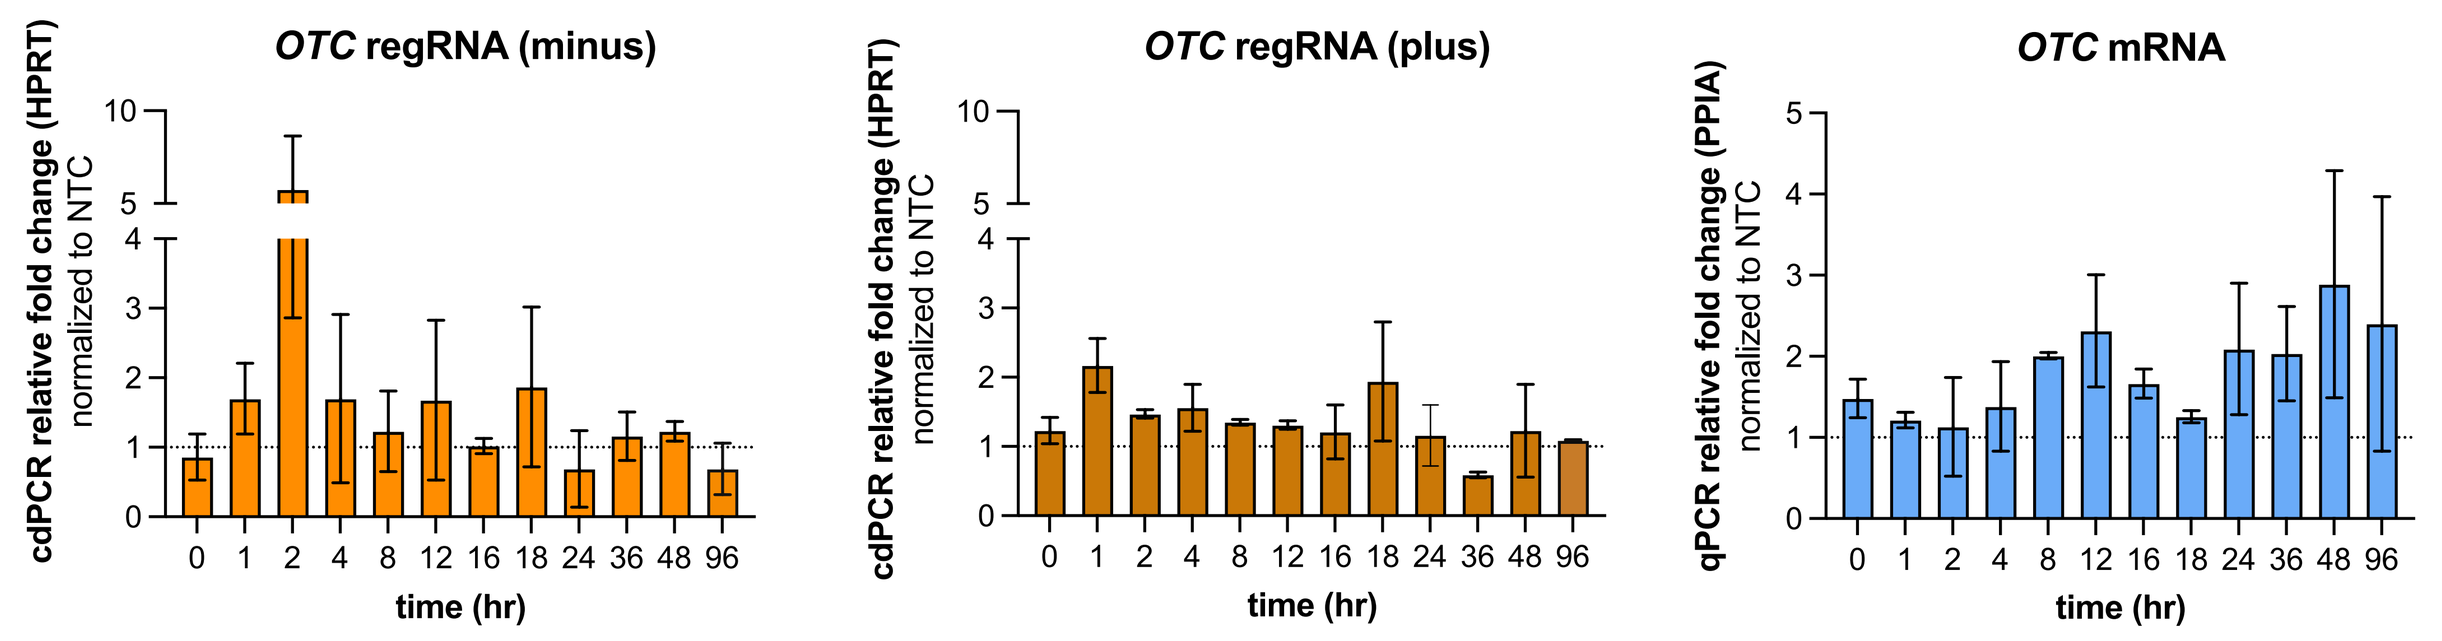
OTC regRNA (minus)
OTC regRNA (plus)
OTC mRNA
cdPCR relative fold change (HPRT) normalized to NTC
qPCR relative fold change (PPIA) normalized to NTC
0 1 2 4 8 12 16 18 24 36 48 96
time (hr)

OTC regRNA-targeting ASOs: mechanism of action

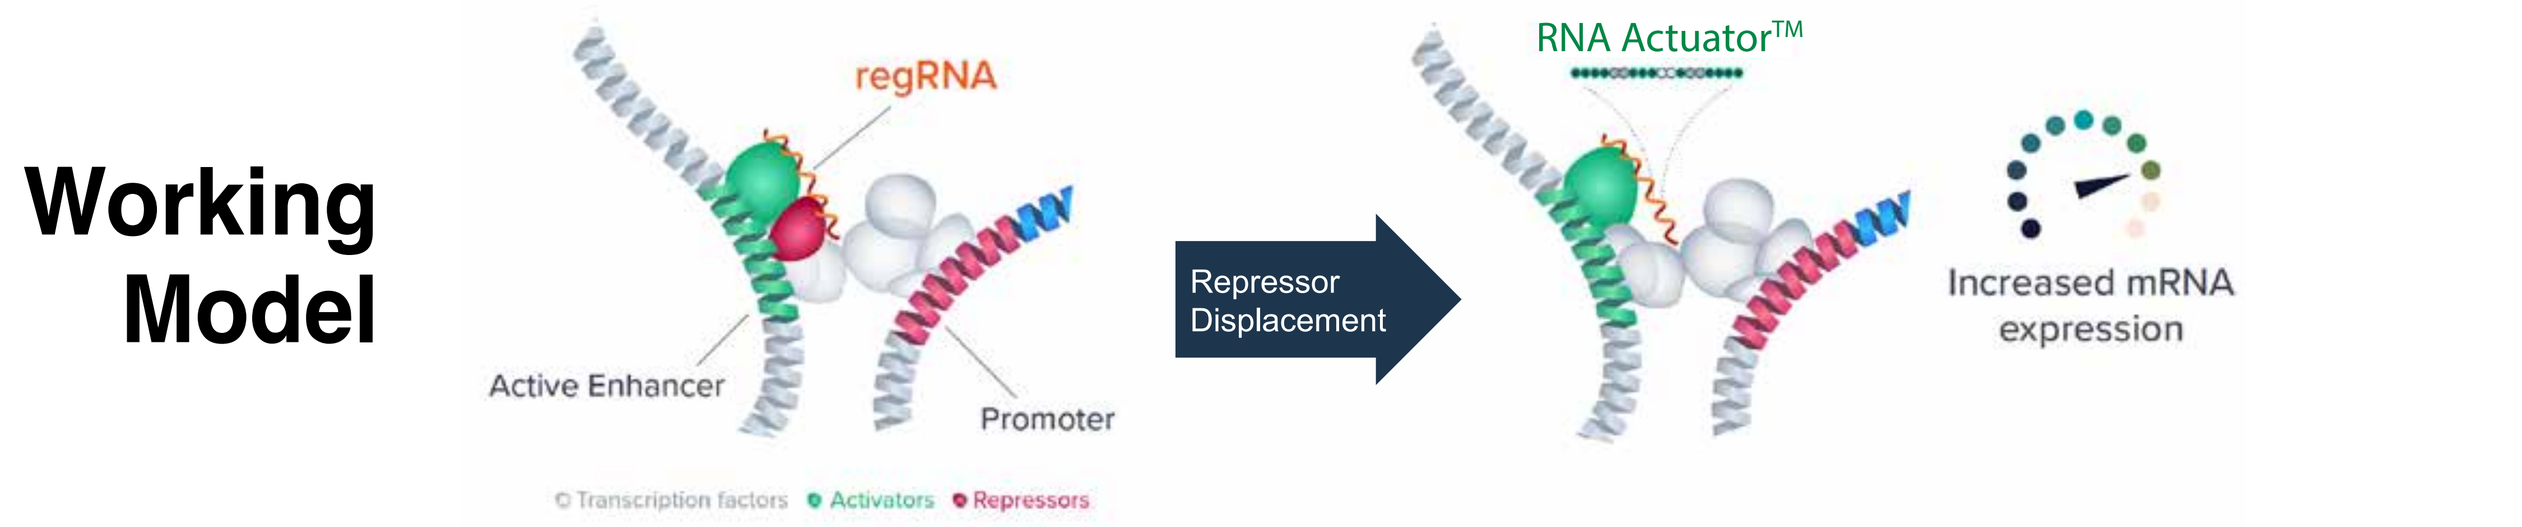
Working Model
regRNA
Active Enhancer
Promoter
Transcription factors
Activators
Repressors
Repressor Displacement
RNA Actuator™
Increased mRNA expression

Treatment with ASO reduces repressor binding at enhancer

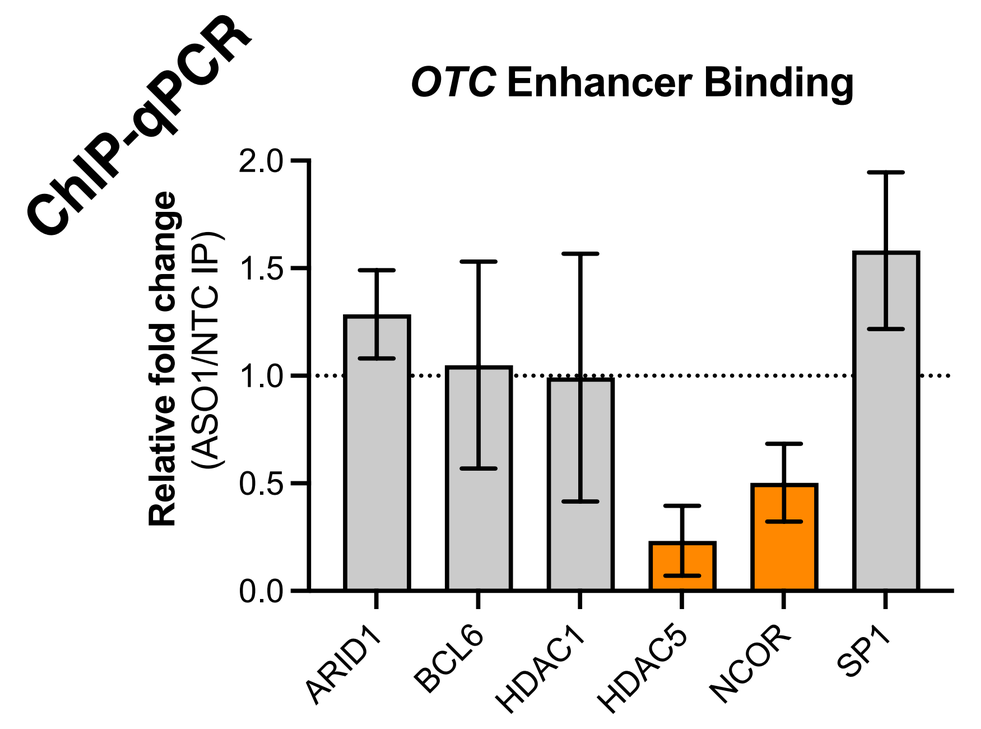
ChIP-qPCR
OTC Enhancer Binding
Relative fold change (ASO1/NTC IP)
ARID1
BCL6
HDAC1
HDAC5
NCOR
SP1

Repressor knockdown results in loss of ASO effect

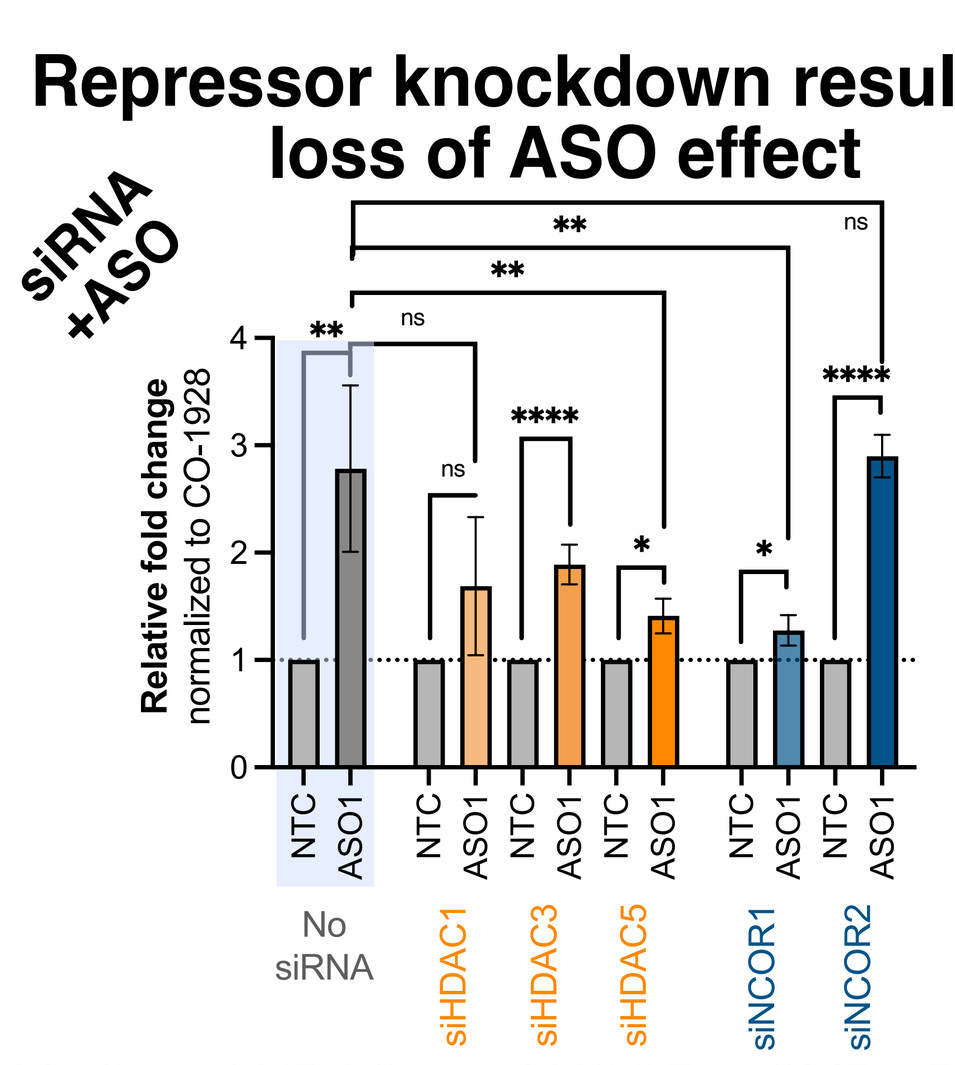
siRNA +ASO
Relative fold change normalized to CO-1928
NTC
ASO1
No siRNA
siHDAC1
siHDAC3
siHDAC5
siNCOR1
siNCOR2
ns
**

*

Left: Treatment with targeting ASO1 results in a decrease in binding of HDAC5 and NCOR1 (ChIP-qPCR)
Right: Knockdown of HDAC1,3,5 or NCOR1 prior to ASO treatment (48h) ablates ASO1 effect on OTC mRNA

ASO binding results in reproducible changes to regRNA structure

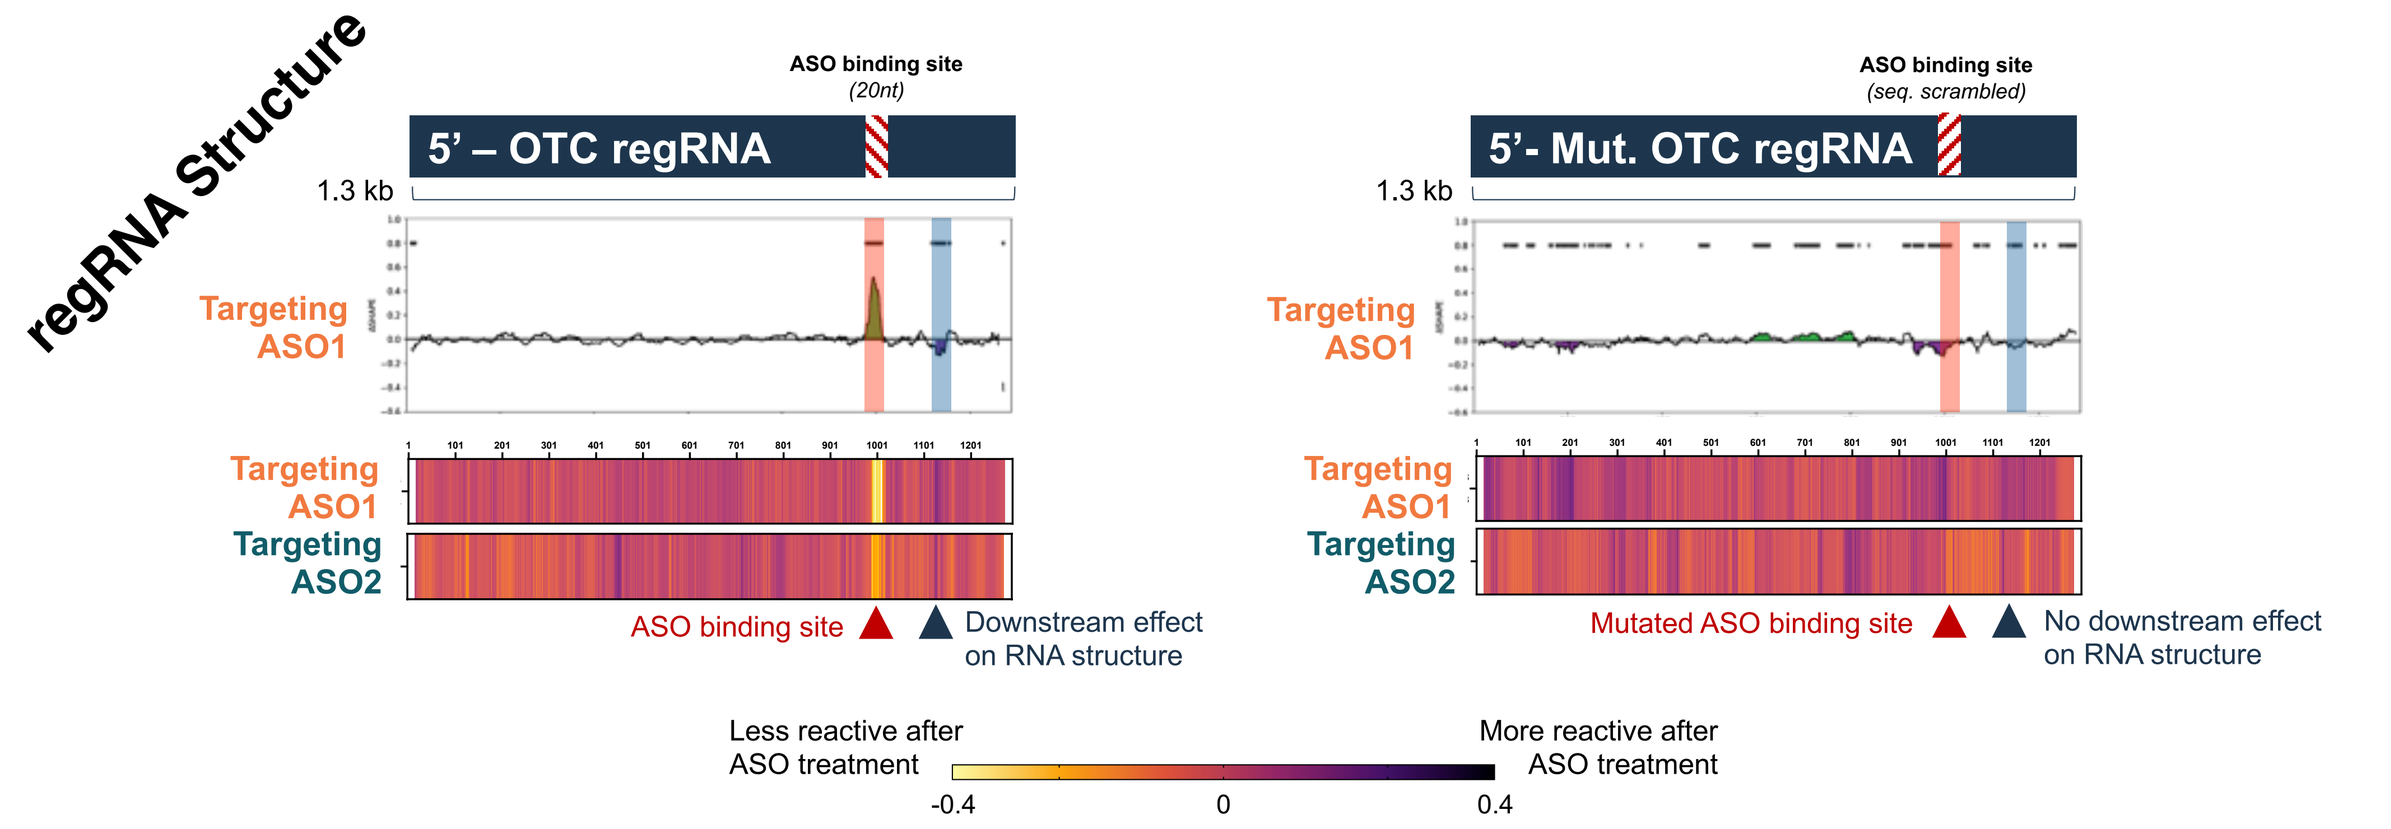
regRNA Structure
ASO binding site (20nt)
5' – OTC regRNA
ASO binding site (seq. scrambled)
5'- Mut. OTC regRNA
1.3 kb
Targeting ASO1
Targeting ASO2
ASO binding site
Downstream effect on RNA structure
Mutated ASO binding site
No downstream effect on RNA structure
Less reactive after ASO treatment
More reactive after ASO treatment
-0.4
0
0.4

Targeting ASO1 binding alters regRNA secondary structure as measured by SHAPE-MaP (RNA nucleotide reactivity relative to an NTC ASO). Effect was not observed for mutant regRNA lacking the ASO binding site (i.e., ASO2).

In vivo efficacy of *OTC* regRNA-targeting ASOs

Humanized liver mouse model allows testing of hOTC-targeting ASOs *in vivo*

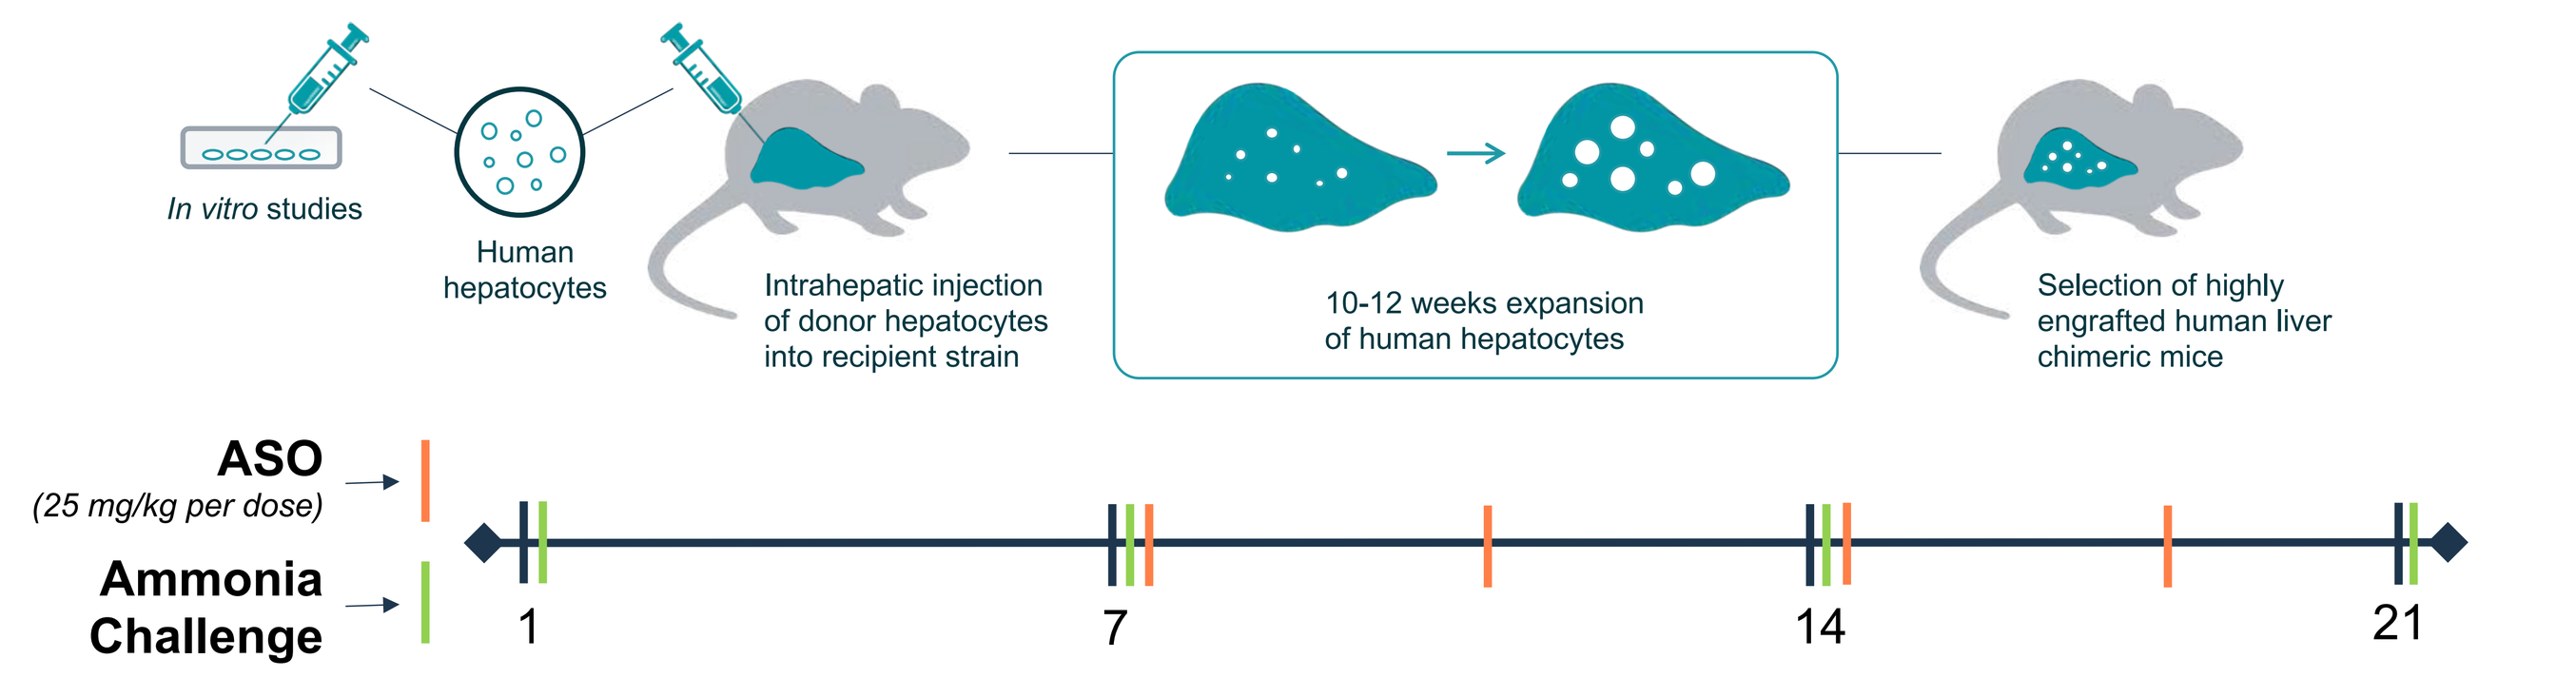
In vitro studies
Human hepatocytes
Intrahepatic injection of donor hepatocytes into recipient strain
10-12 weeks expansion of human hepatocytes
Selection of highly engrafted human liver chimeric mice
ASO
(25 mg/kg per dose)
Ammonia Challenge
1
7
14
21

regRNA-targeting ASO leads to increased OTC protein and ureagenesis

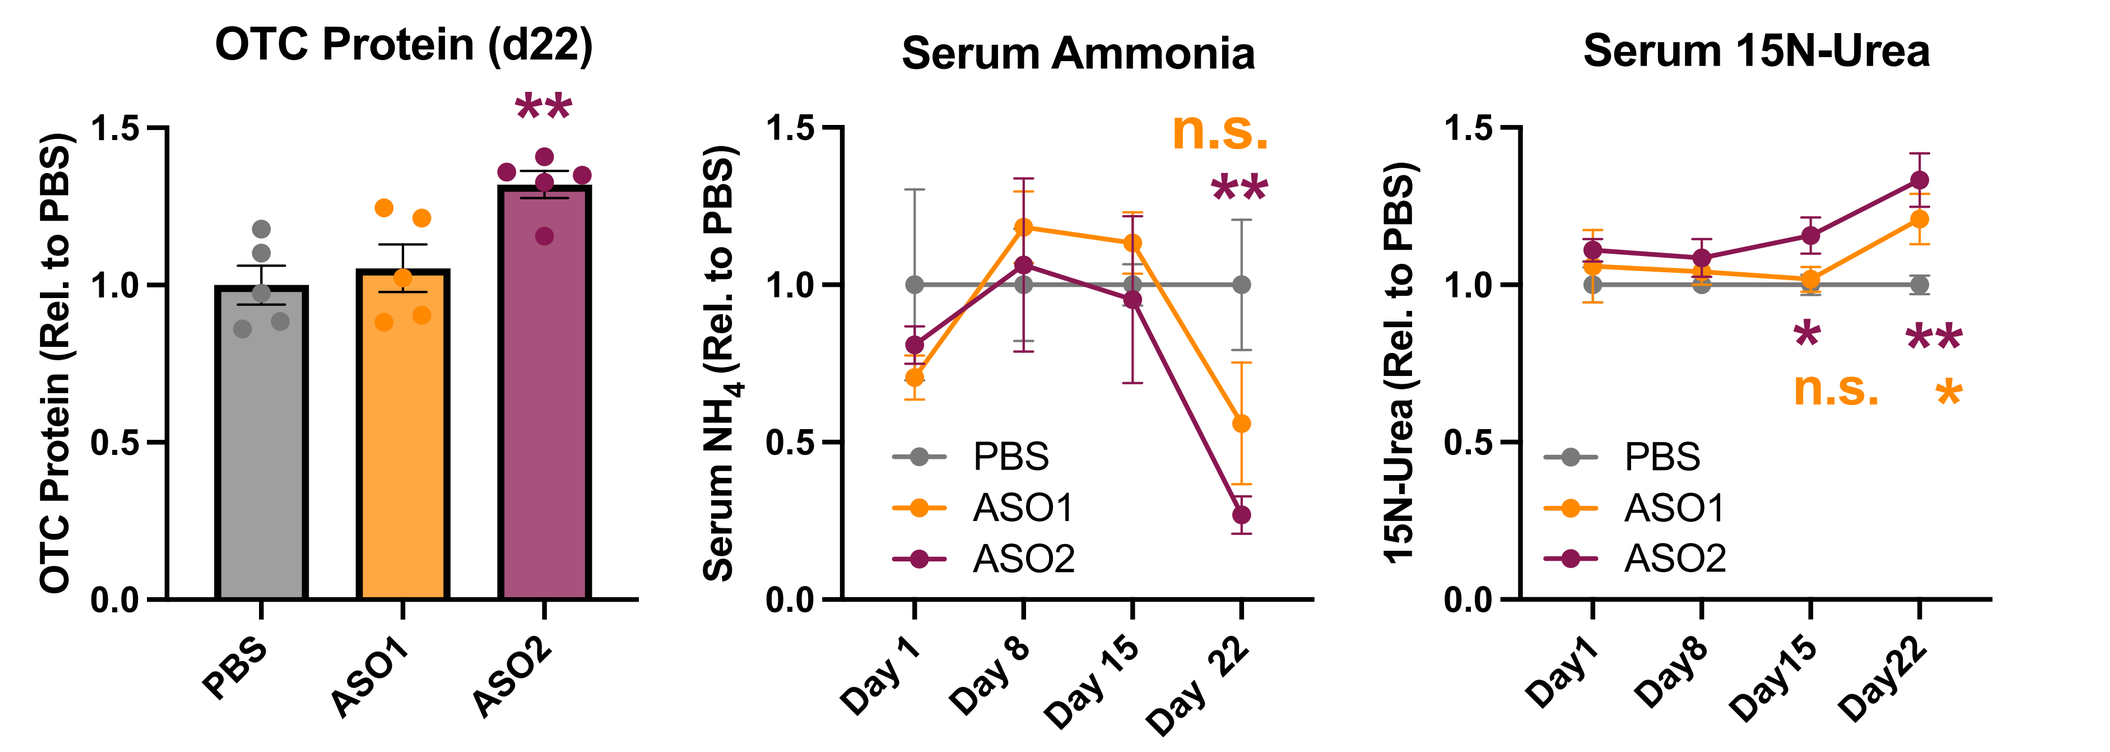
OTC Protein (d22)
Serum Ammonia
Serum 15N-Urea
OTC Protein (Rel. to PBS)
Serum NH4 (Rel. to PBS)
15N-Urea (Rel. to PBS)
PBS
ASO1
ASO2
n.s.
**
*
Day 1
Day 8
Day 15
Day 22

Left: OTC protein levels were measured at final endpoint, day 22, using capillary-based Western.
Center: Serum ammonia was measured from fresh serum at the indicated day. Data is normalized to the matched timepoint PBS control +/- SEM for n=5 mice per group.
Right: Labeled serum urea following a challenge with 15N-NH_4 was measured by LC-MS/MS. Data is normalized to the matched timepoint PBS. P values are one way ANOVA. * p<0.05, **p<0.01

Takeaways

ASOs targeting *OTC* regRNAs upregulate *OTC* mRNA in a dose-dependent

OTC regRNA upregulation preceded mRNA upregulation after ASO treatment

ASOs modulate repressor binding leading to increased *OTC* transcription

ASOs targeting *OTC* regRNAs increase OTC protein levels and decrease ammonia levels in humanized liver mouse model

CAMP4's RAP Platform™ facilitates the development of precise, potent therapeutic candidates that can be programmed to treat a broad range of diseases